Estimation of occupancy and radioligand kinetics in the CNS from PET-data in the absence of a reference region

M. Kågedal(1,3), Z. Cseleny(1), S. Nyberg(1), Pär Karlsson (2), A Hooker(3), MO Karlsson(3)
(1) AstraZeneca R&D Södertälje, Sweden
(2) Karolinska institutet, Department of Clinical neuroscience, Psychiatry Section, Karolinska Hospital, Stockholm, Sweden
(3) Department of Pharmaceutical Biosciences, Uppsala University, Uppsala, Sweden

1743

Objectives:
With traditional methods calculation of occupancy is based on the specific uptake (binding potential) estimated individually from each PET-measurement. Estimation of occupancy is not possible with this approach when a reference region void of receptors is lacking. In order to estimate the *in vivo* affinity (Kd_{pl}), a population model was developed for receptor binding kinetics by use of measured concentrations of ^{11}C-AZDX in plasma and CNS as well as concentrations of unlabelled AZDX in plasma.

Methods:
The analysis was based on data from a PET study in six healthy volunteers. On four occasions each subject was examined by a 60-90 minute PET measurement after injection of tracer amount of the radioligand. The radioligand (^{11}C-AZDX) was given alone on one occasion and at 3 hours after oral administration of different doses of unlabelled AZDX on the other occasions. The time course of radioligand concentration in regions of interest in the CNS was derived from the PET-measurements and the time course of unchanged drug in plasma was derived from measurements in arterial blood and plasma.

In order to improve the ability to separate between receptor bound and non-specific uptake in the CNS, the two regions with the highest (Ventral striatum, VST) and lowest (cerebellum, CER) uptake were included in the analysis. Differential equations reflecting the model illustrated in Figure 1 were used to estimate the transfer between plasma and CNS and the binding to the receptor. It was assumed that the extent of non-displaceable uptake (KP_{ND}=K1/k2) as well as the receptor binding kinetics were the same in both regions while the rate of CNS uptake and the receptor density (Bmax) could differ. The observed concentration in the brain (C_{CER}, C_{VST}) corresponded to the sum of non-displaceable (C_{ND}), receptor bound (C_B) and the contribution from blood. The model accounted for the difference in residual error in the regions (ε_{CER}, ε_{VST}) as well as the correlation between them (ε_{joint}) (Eq1,2). The relationship between free receptor concentration (R_{Free}) and drug concentration in the brain was included as a saturation model (Eq 3).

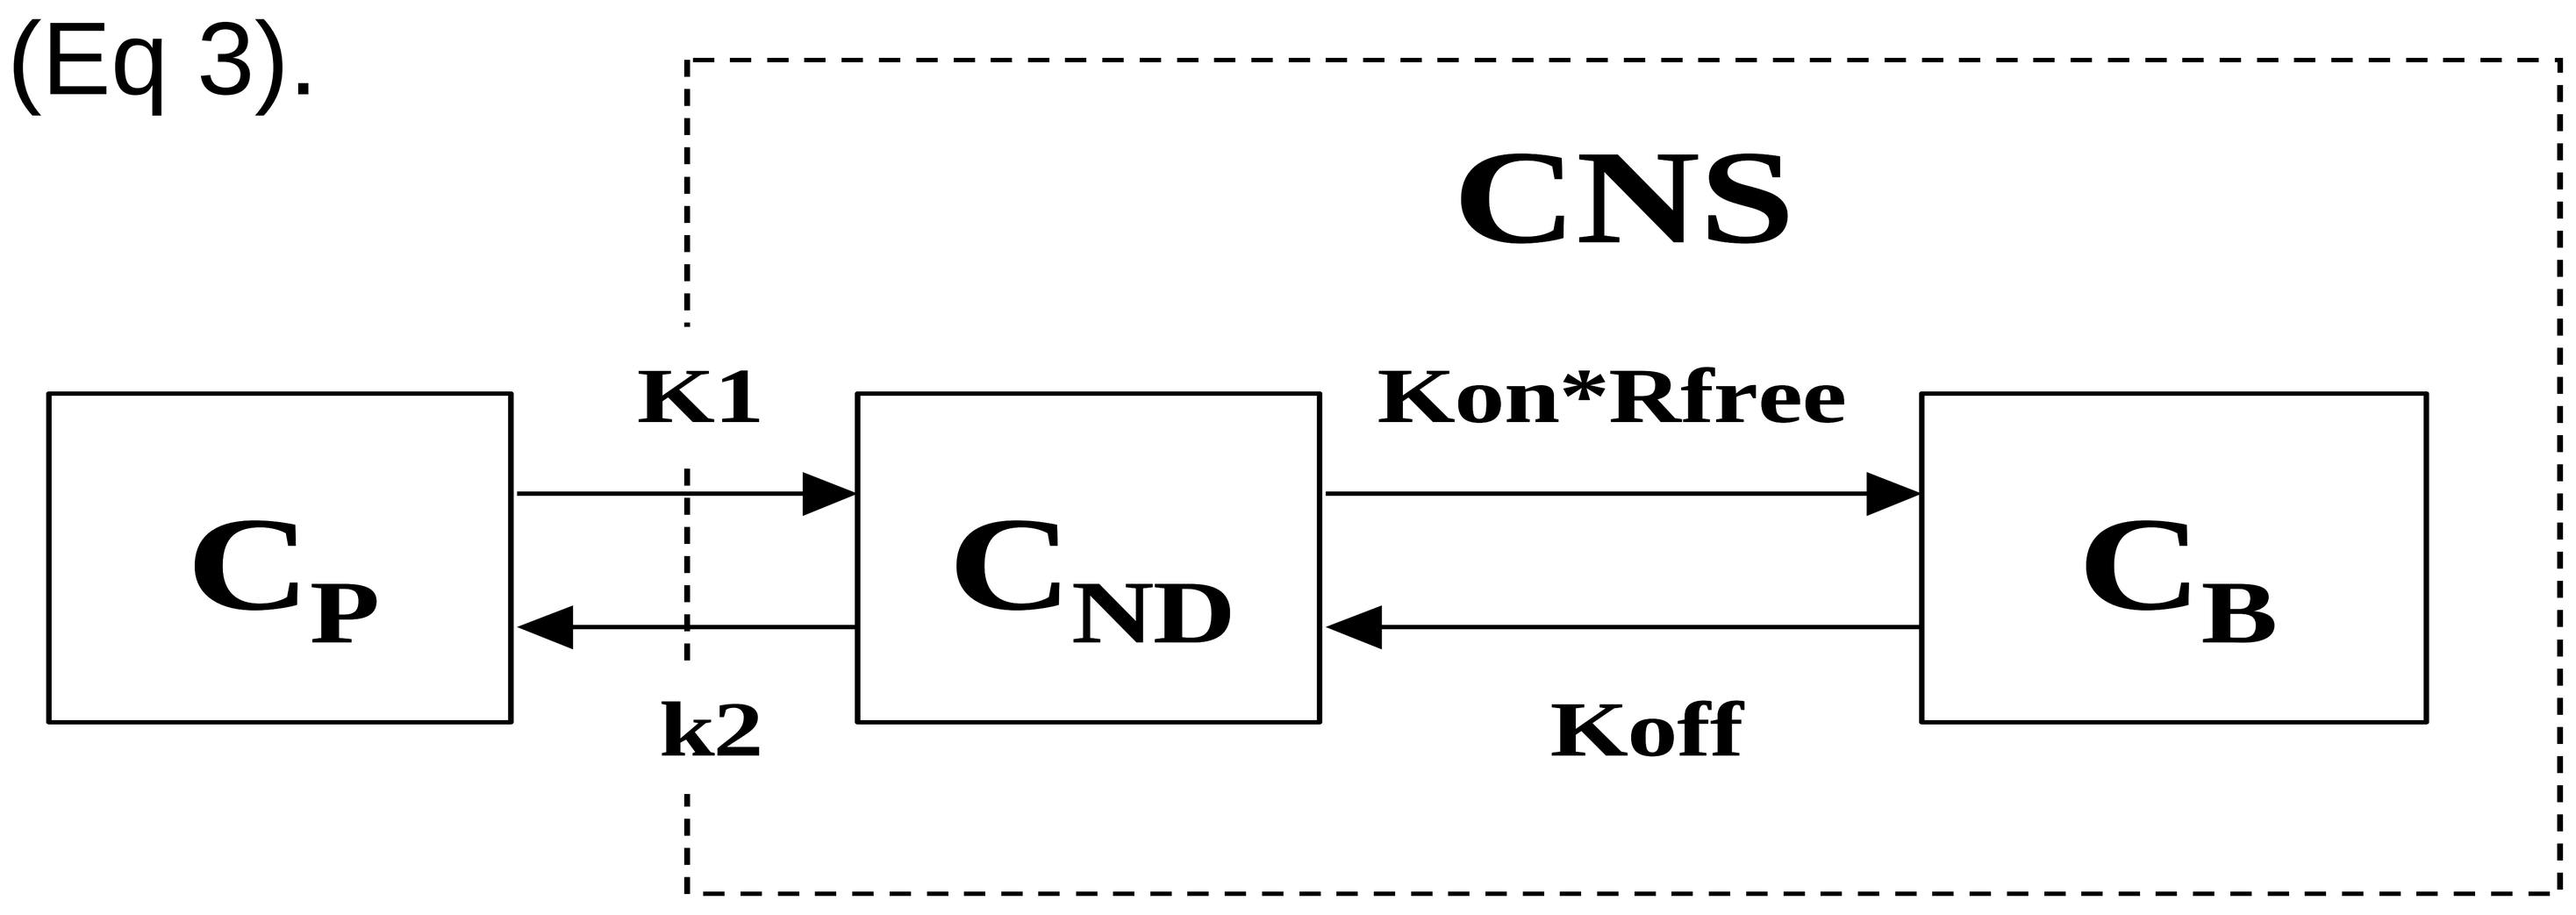
CNS
C_P
C_{ND}
C_B
K1
k2
Kon*Rfree
Koff

Figure 1. The model included the exchange between plasma and brain as well as the binding to receptors in the brain.

Results:
- AZDX binding at the receptor is saturable with an estimated Kd_{pl} of 200nmol/L (RSE=13%). (With only VST data included the RSE was 84%.)
- The density of the receptor binding sites are approximately 800nM and 200nM in VST and CER respectively.

Equations:

$$C_{CER}=(C_{ND,CER}+C_{B,CER})*(1-RBV)+(Cbl*RBV)+\varepsilon_{CER}+\varepsilon\, joint \quad \text{(Eq. 1)}$$

$$C_{VST}=(C_{ND,VST}+C_{B,VST})*(1-RBV)+(Cbl*RBV)+\varepsilon_{VST}+\varepsilon\, joint \quad \text{(Eq. 2)}$$

$$R_{Free}=B_{MAX}\cdot\left(1-\frac{C_{ND}}{C_{ND}+Kd_{ND}}\right) \quad \text{(Eq. 3)}$$

$$Kd_{pl}=Kd_{ND}/KP_{ND} \quad \text{(Eq. 4)}$$

$$KP_T=\frac{K1}{k2}\cdot\left(1+\frac{Kon\cdot Rfree}{Koff}\right) \quad \text{(Eq. 5)}$$

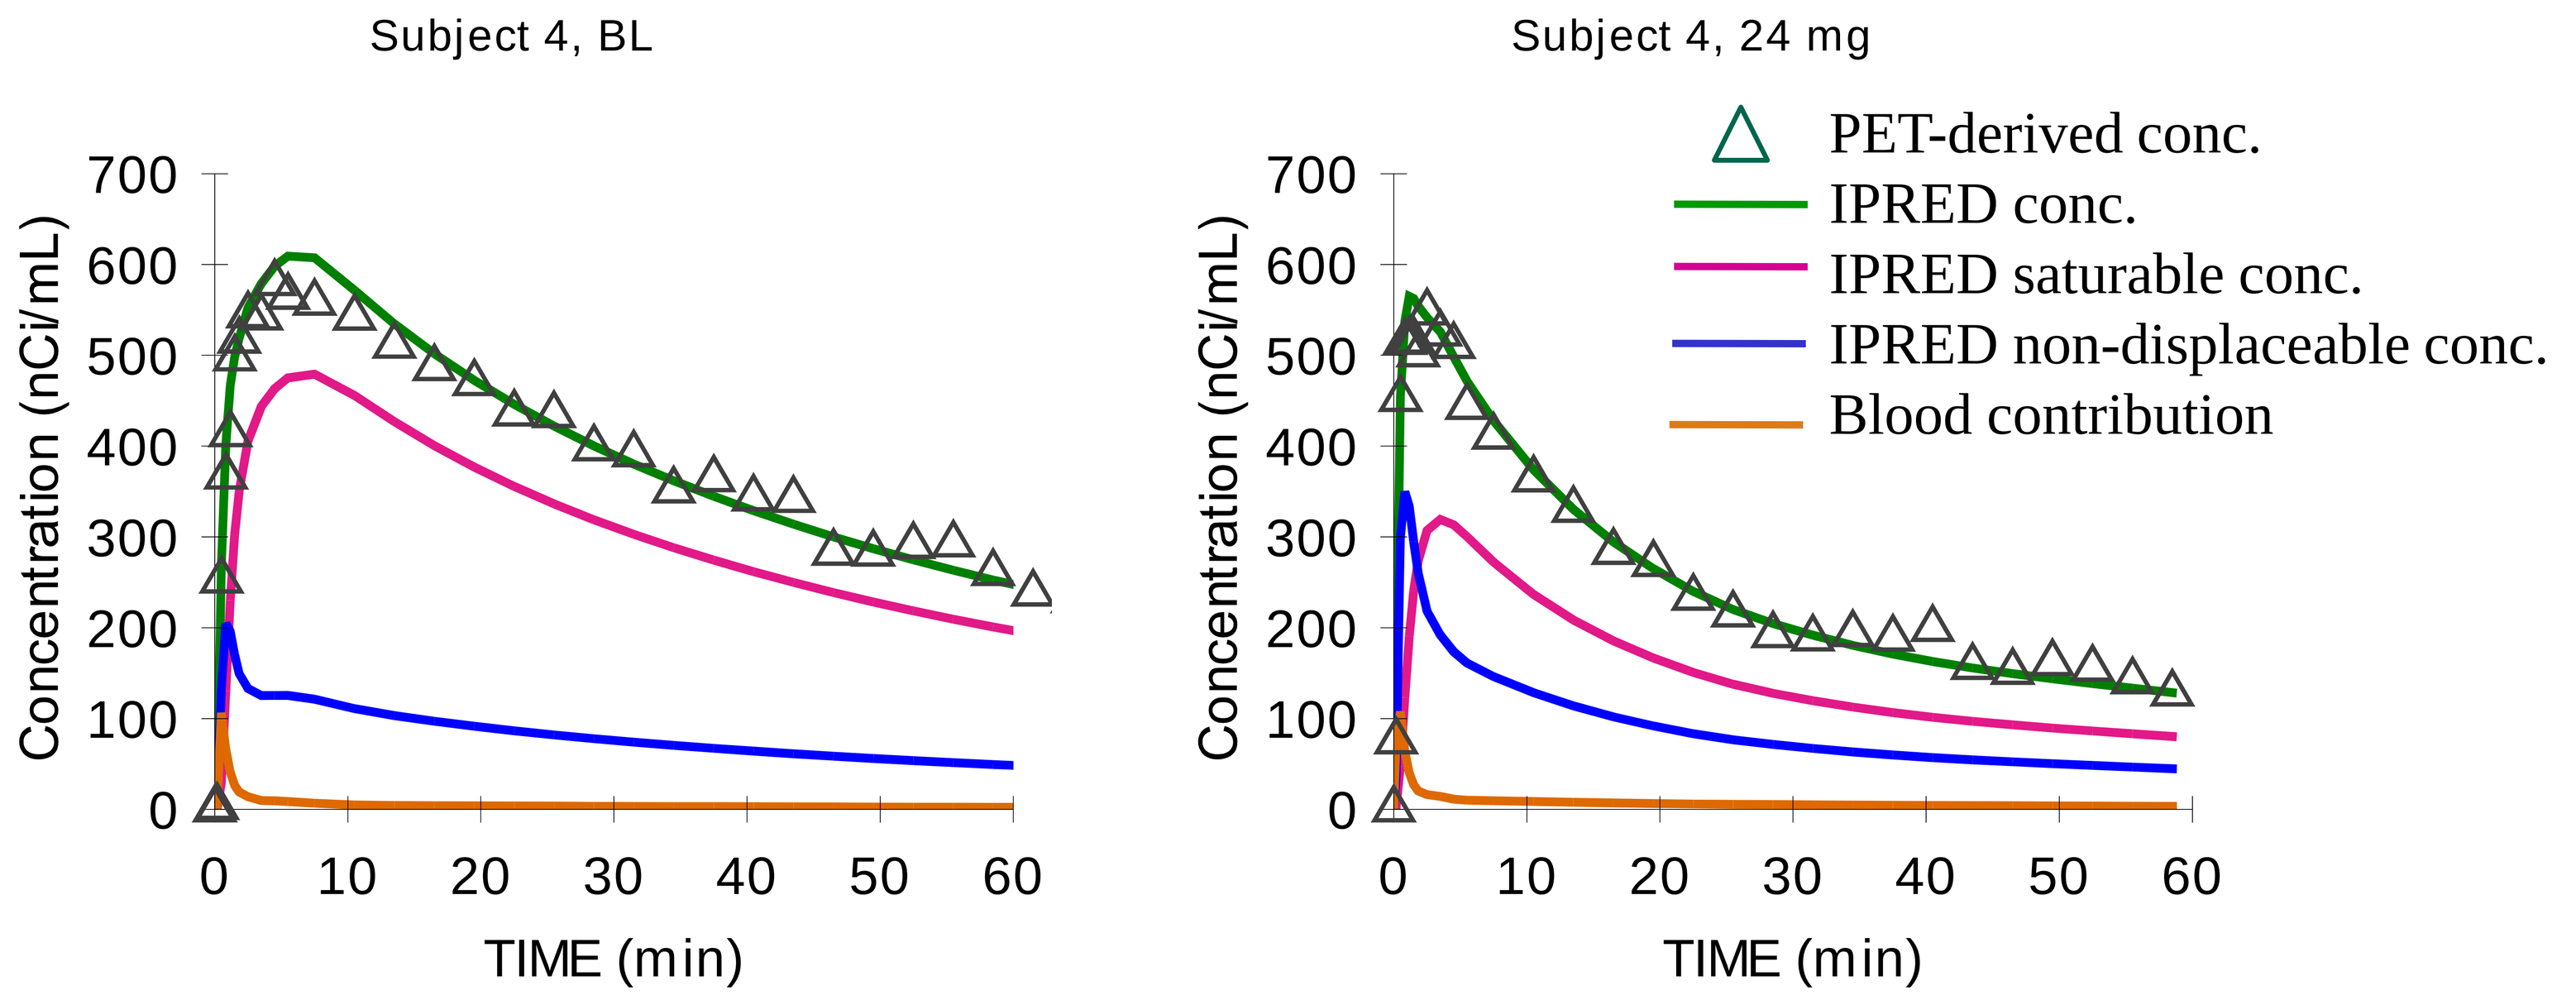
Subject 4, BL
Subject 4, 24 mg
Concentration (nCi/mL)
TIME (min)
PET-derived conc.
IPRED conc.
IPRED saturable conc.
IPRED non-displaceable conc.
Blood contribution

Figure 2. Radioligand concentration in VST versus time at base-line and after pre-treatment with highest dose given.

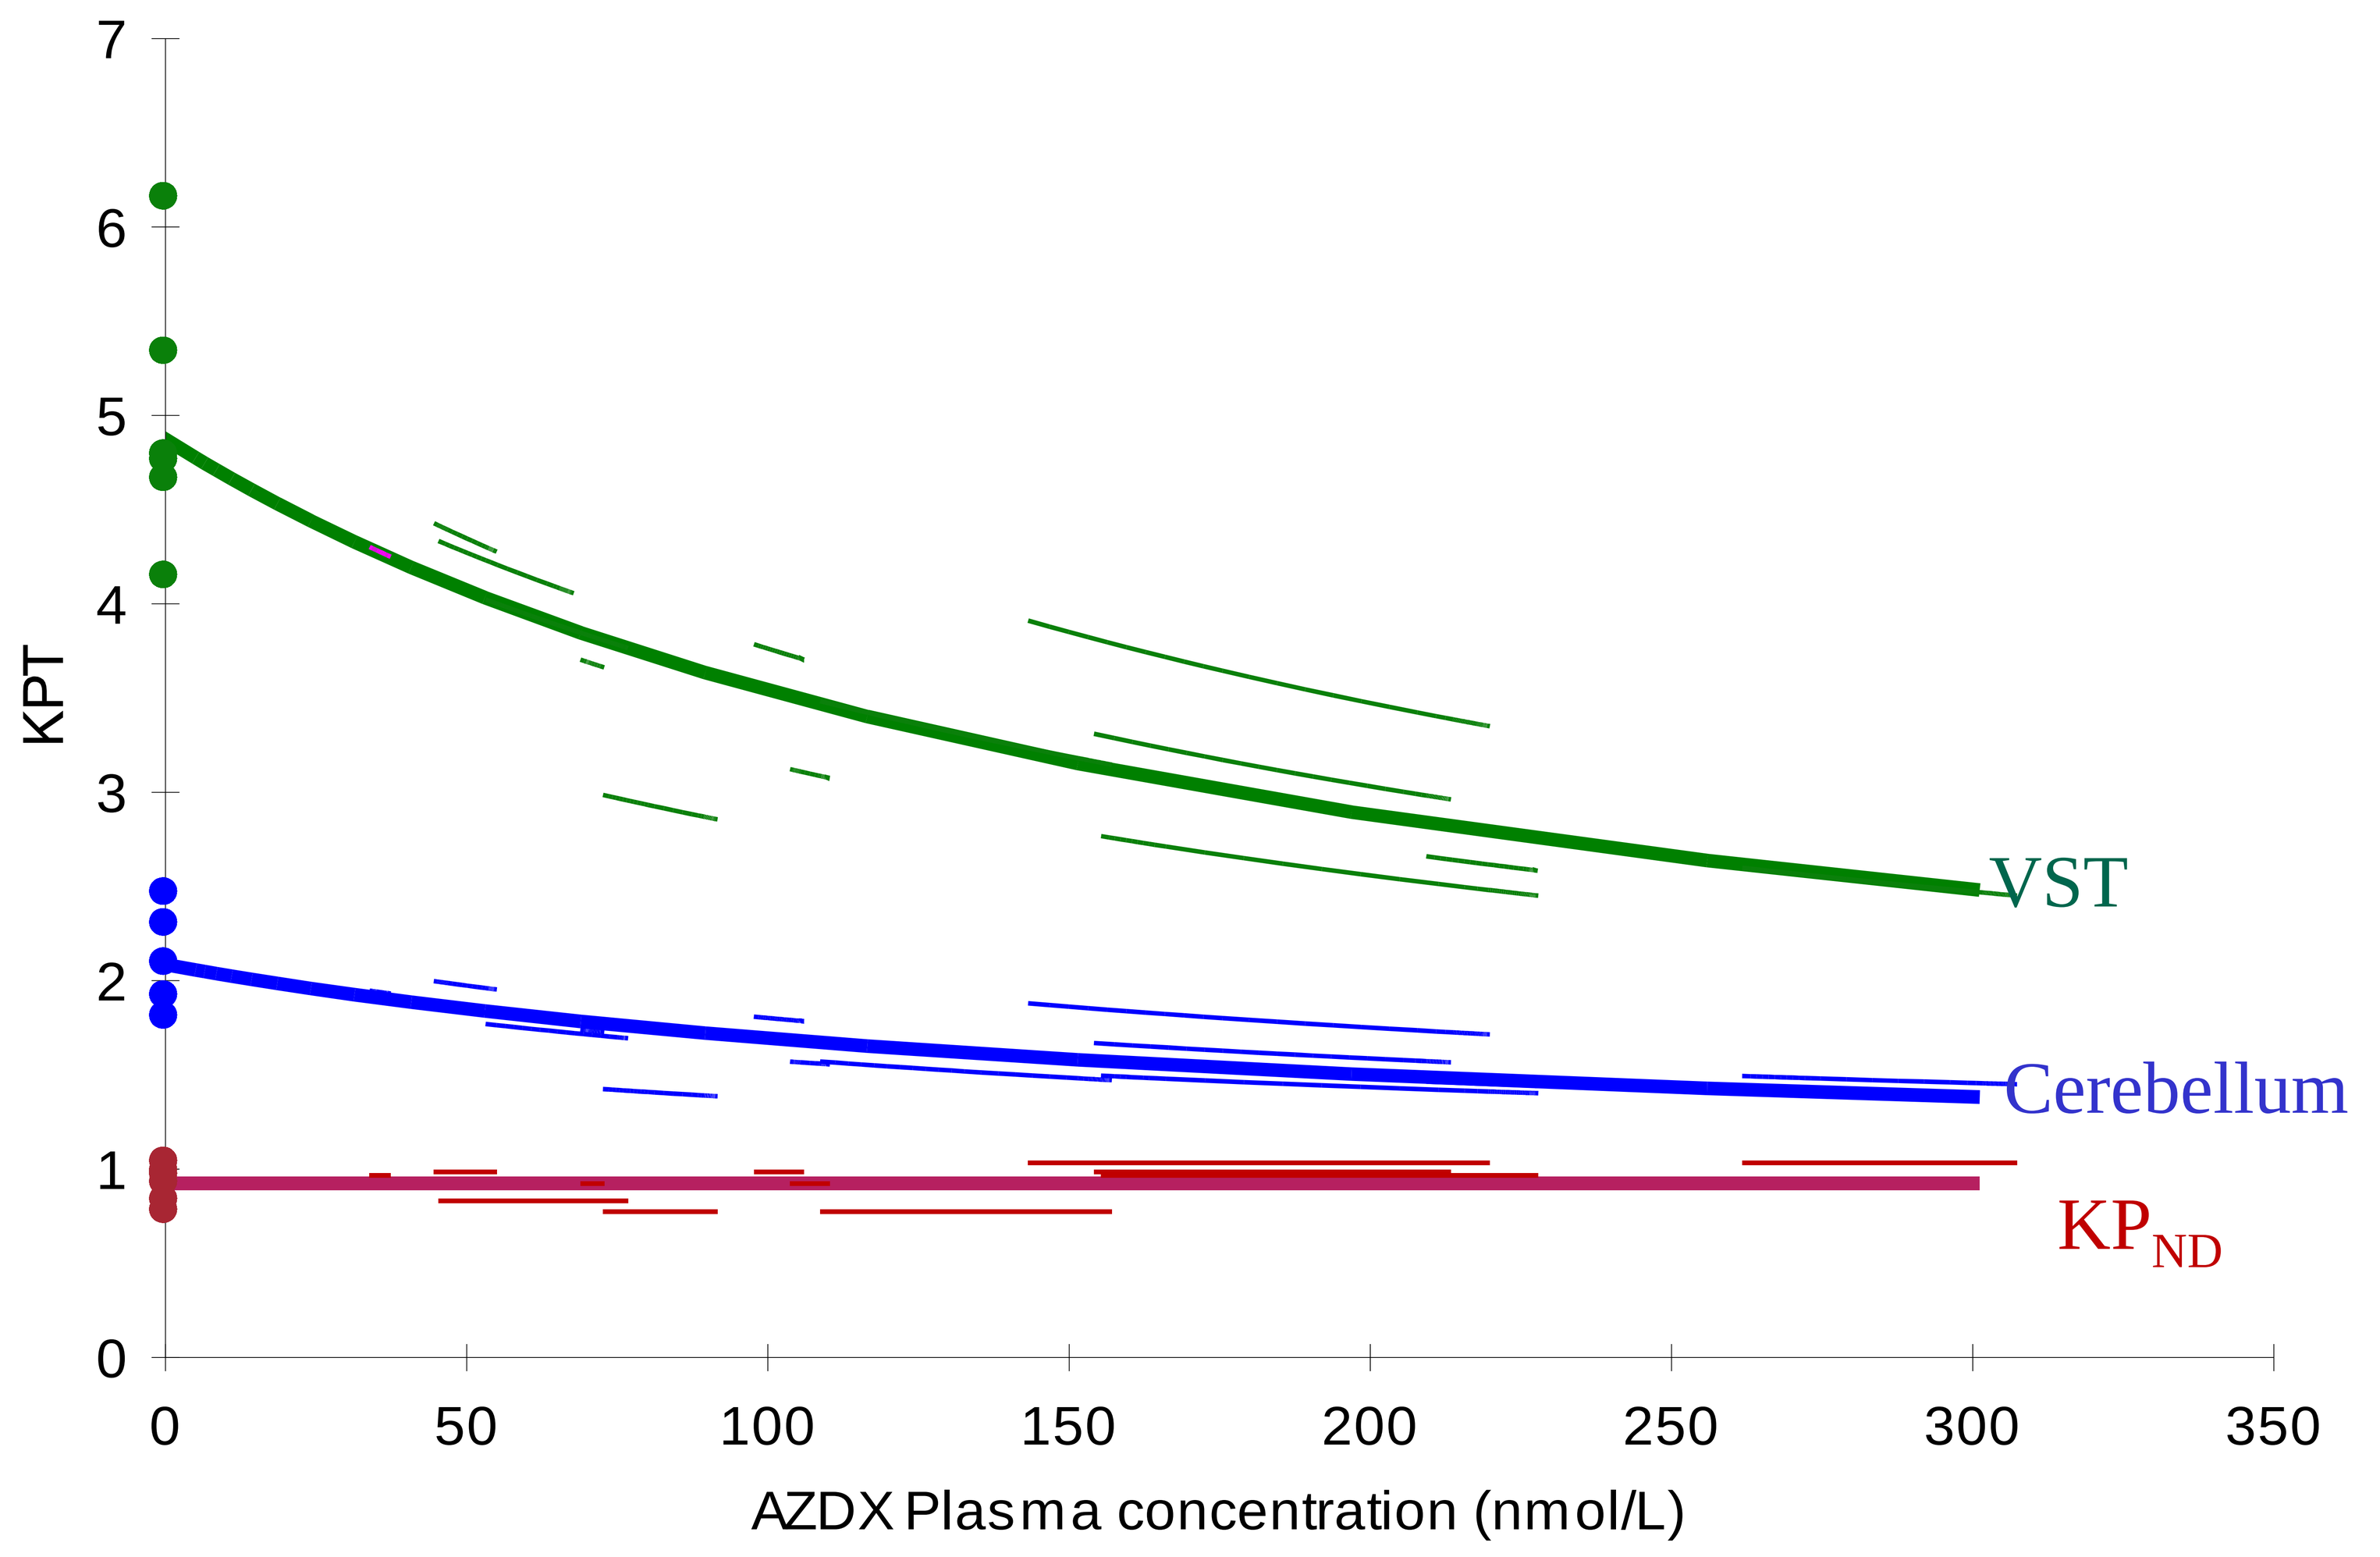
KPT
AZDX Plasma concentration (nmol/L)
VST
Cerebellum
KP_{ND}

Figure 3. Population and individual model predictions of total partition coefficient (KP_T) in VST and CER and KP_{ND} versus plasma concentration.

Conclusions:

By simultaneously analysing data from several PET-measurements in a non-linear mixed effects framework it is possible to estimate parameters of interest that would otherwise be difficult to assess. It is also possible to include a changing cold drug concentration during the PET-assessment rather than having to approximate the concentration with a mean value.